Interactive Voice Response and Tablet Self-Report Versions of the Electronic Columbia-Suicide Severity Rating Scale Are Equivalent

Chad Gwaltney, Ph.D.[1]; Jim Mundt, Ph.D.[2]; John H. Greist MD[3]; Jean Paty, Ph.D.[4], Brian Tiplady, Ph.D.[1], Kelly Posner, Ph.D.[5]

Affiliations:
1 ERT
2 ePRO Research Consulting, LLC
3 Healthcare Technology Systems
4 Quintiles
5 Columbia University

Abstract

Background: Prospective monitoring of suicidal ideation and behavior (SIB) is an FDA requirement in all CNS clinical trials. The interactive voice response (IVR) version of the Electronic Columbia-Suicide Severity Rating Scale (eC-SSRS) – which is completed directly by patients, without a clinician interviewer – can be used to prospectively monitor SIB.

Aims: In this randomized, crossover study, we examined the equivalence of scores obtained from the IVR version and a new patient-reported version that was administered on a tablet device.

Method: Two groups were included in the study - recently admitted psychiatric inpatients (n=58) and employees of the hospital site (n=28) - in order to explore the broadest range of SIB. Participants completed both the IVR and tablet versions in randomized order, with a 25-minute break between administrations. Participants also completed a second administration of the first administered version at the end of the session. Intraclass correlation coefficients (ICCs) and Kappa coefficients were used to evaluate degree of agreement across modalities.

Results: The mean age of the sample was 41.0 years (SD = 12.5); 59% were female. High levels of agreement were observed for most severe lifetime (ICC=0.88) and recent (ICC=0.79) ideation, occurrence of actual lifetime (Kappa=0.81) and recent (Kappa = 0.73) suicide attempts, and occurrence of lifetime interrupted attempts (Kappa=0.78), aborted attempts (kappa=0.54), and preparatory behaviors (Kappa=0.77), as well as non-suicidal self-injurious behavior (Kappa=0.73). Scores from both modes significantly differentiated psychiatric patients and hospital employee controls and the test-retest reliability of both modes was excellent.

Conclusions: These results support the validity and reliability of the new tablet-based eC-SSRS. This will allow the inclusion of the eC-SSRS in a wider range of clinical studies, particularly where a tablet is also being used to collect other patient-reported efficacy or safety data.

FDA and Suicidal Ideation and Behavior

Prospective monitoring of suicidal ideation and behavior (SIB) is recommended by FDA (FDA, 2012) in several therapeutic areas, including all clinical trials of psychiatry and neurology products

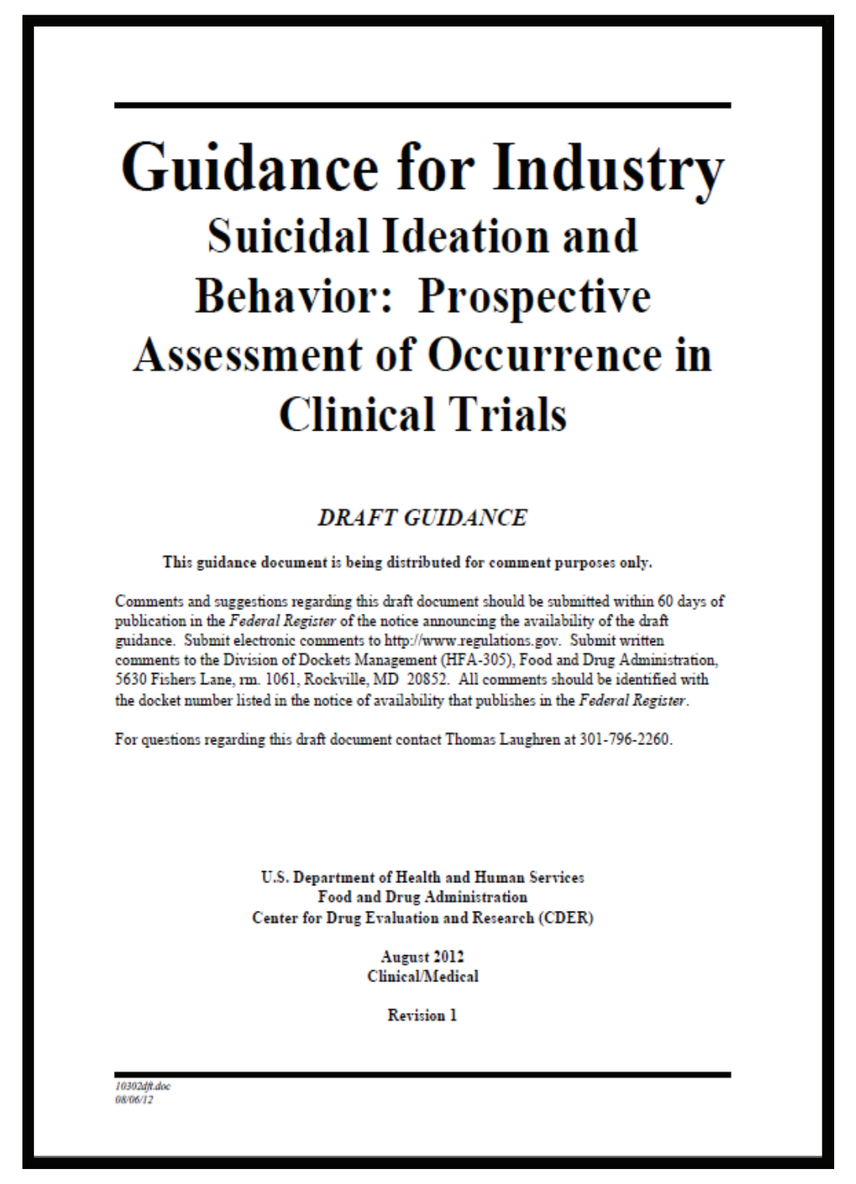
Guidance for Industry
Suicidal Ideation and Behavior: Prospective Assessment of Occurrence in Clinical Trials

DRAFT GUIDANCE

This guidance document is being distributed for comment purposes only.

Comments and suggestions regarding this draft document should be submitted within 60 days of publication in the *Federal Register* of the notice announcing the availability of the draft guidance. Submit electronic comments to http://www.regulations.gov. Submit written comments to the Division of Dockets Management (HFA-305), Food and Drug Administration, 5630 Fishers Lane, rm. 1061, Rockville, MD 20852. All comments should be identified with the docket number listed in the notice of availability that publishes in the *Federal Register*.

For questions regarding this draft document contact Thomas Laughren at 301-796-2260.

U.S. Department of Health and Human Services
Food and Drug Administration
Center for Drug Evaluation and Research (CDER)

August 2012
Clinical/Medical

Revision 1

Columbia-Suicide Severity Rating Scale (C-SSRS)

- The C-SSRS, a clinician-administered measure, is an "acceptable" (FDA, 2012) method for assessing SIB
- An electronic version of the C-SSRS – the eC-SSRS – has been developed to directly collect patient reports of SIB and increase operational efficiency
- Originally developed for administration by Interactive Voice Response (IVR) system

C-SSRS Implementation Options

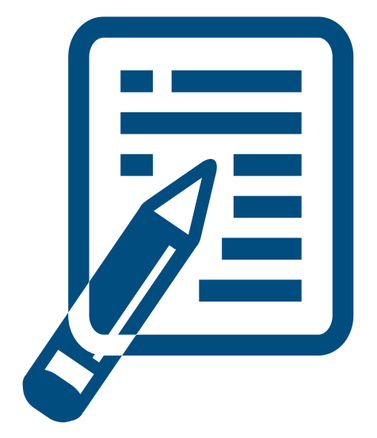

Paper C-SSRS

ClinRO
Semi-structured
Paper
Data available after data entry
Clinician time to administer
Many queries about paper forms

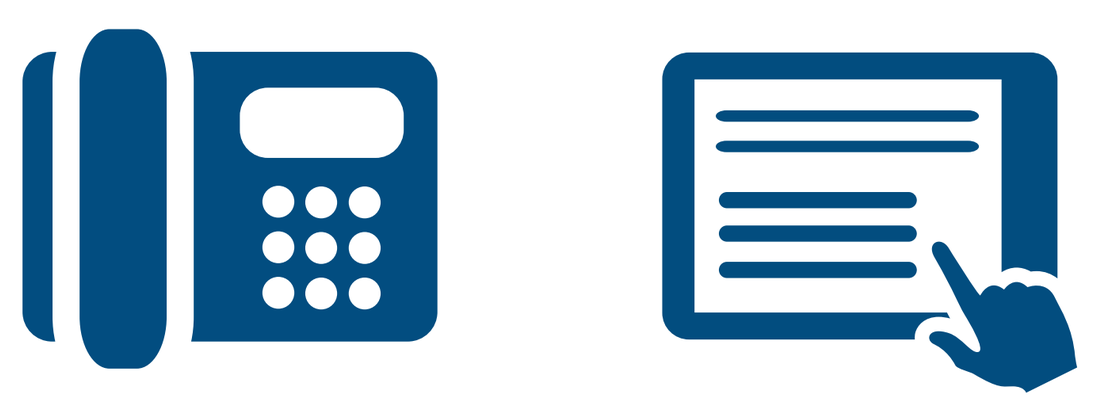

Tablet & IVR eC-SSRS

PRO
Fully-structured
Interactive voice response
Touch screen tablet
Data available immediately
Clinician workload dramatically reduced
Queries dramatically reduced

Goals of Current Study

To assess the equivalence of the subject-reported scores obtained by a novel electronic tablet version and IVR administration of the C-SSRS.

To assess the within-modality test-retest reliability of the eC-SSRS versions.

Study Design

Randomized Crossover Design

	Sequence A	Sequence B
Period 1	Tablet	IVR
5 minute distractor task followed by 20 min refreshment break		
Period 2	IVR	Tablet
5 minute distractor task followed by 20 min refreshment break		
Period 3	Tablet	IVR
	Technology / Ease of Use / Preference scale	

Recruitment

- Inpatient Psychiatric Patients
 - Recruited by site staff doing chart review
 - If a patient appeared eligible, staff requested that the attending doctor allow study staff to approach the patient regarding participation
- Hospital Workers
 - Recruited through flyers in Rogers Memorial Hospital (WI)
 - None of the staff had experience with the C-SSRS or eC-SSRS
- Study coordinator met with the participants to describe the study details, answer any questions and concerns
- Procedures approved by IRB

Assessments

- eC-SSRS: The tablet and IVR versions (provided by ERT, inc.) are derived from the baseline/lifetime version of the scale with recency assessment of:
 - 6 months for suicidal ideation
 - 2 years for suicidal behavior

Scoring

Suicidal Ideation
1. Passive
2. Active: Nonspecific, no method, intent, or plan
3. Active: Method, but no intent or plan
4. Active: Method and intent, but no plan
5. Active: Method, intent, and plan

Suicidal Behavior
1. Completed suicide (N/A in this study)
2. Suicide attempt
3. Interrupted attempt
4. Aborted attempt
5. Preparatory actions toward imminent suicidal behaviors
 Nonsuicidal, self-injurious behavior

- Scores used in this study
 - Most severe ideation lifetime (0-5)
 - Most severe ideation in past 6 months (0-5)
 - Lifetime presence of each type of behavior (Yes/No)
 - Presence of each type of behavior in past 2 years (Yes/No)
 - Number of lifetime suicide attempts (continuous variable)

Analysis Plan

- Primary measures of agreement:
 - Kendall's Tau-b (ideation)
 - Intraclass correlation coefficient (ICC) calculated using the form ICC (2,1) (ideation)
 - Kappa coefficients (for binary behavior assessments)
- Statistics calculated between the first and second assessments (for inter-mode agreement) and between the first and third assessments (test-retest agreement; ideation only)
- The target value for good agreement is ≥ 0.5 for Tau-b, ≥ 0.7 for ICC, and ≥ 0.6 for kappa
- Comparison of patient and control groups using data from 1st administration of eC-SSRS (known-groups validity)
 - ANOVA or chi-square analysis

Demographics

Characteristic	Inpatient Psychiatric Patients (n=58)	Hospital Worker Controls (n=28)	Total Sample (N=86)
Gender	52% Female	82% Female**	59% Female
Age	M = 40.1; SD 12.7	M= 43.1; SD 12.0	41 Years; SD = 12.5
Race			
African American	9	8	17
Caucasian	45	18	63
Latino	4	0	4
Nigerian	0	1	1
Nigerian-American	0	1	1

**There was a significantly higher proportion of females in the control group than in the patient sample; $X^2(1) = 8.97$, p = 0.003.

Convergence Analyses - Ideation

- Measures of equivalence all above threshold for convergence
- Test-retest reliability is high for both measures

Variable	Equivalence Tau-b	Equivalence ICC	Test-restest IVR Tau-b	Test-retest IVR ICC	Test-retest Tablet Tau-b	Test-retest Tablet ICC
Most Severe Lifetime Ideation	.87, p<.001	.89, p<.001	.82, p<.001	.87, p<.001	.82, p<.001	.87, p<.001
Most Severe Recent (last 6 months) Ideation	.69, p<.001	.79, p<.001	.63, p<.001	.72, p<.001	.76, p<.001	.84, p<.001

Known-Groups Validity - Ideation

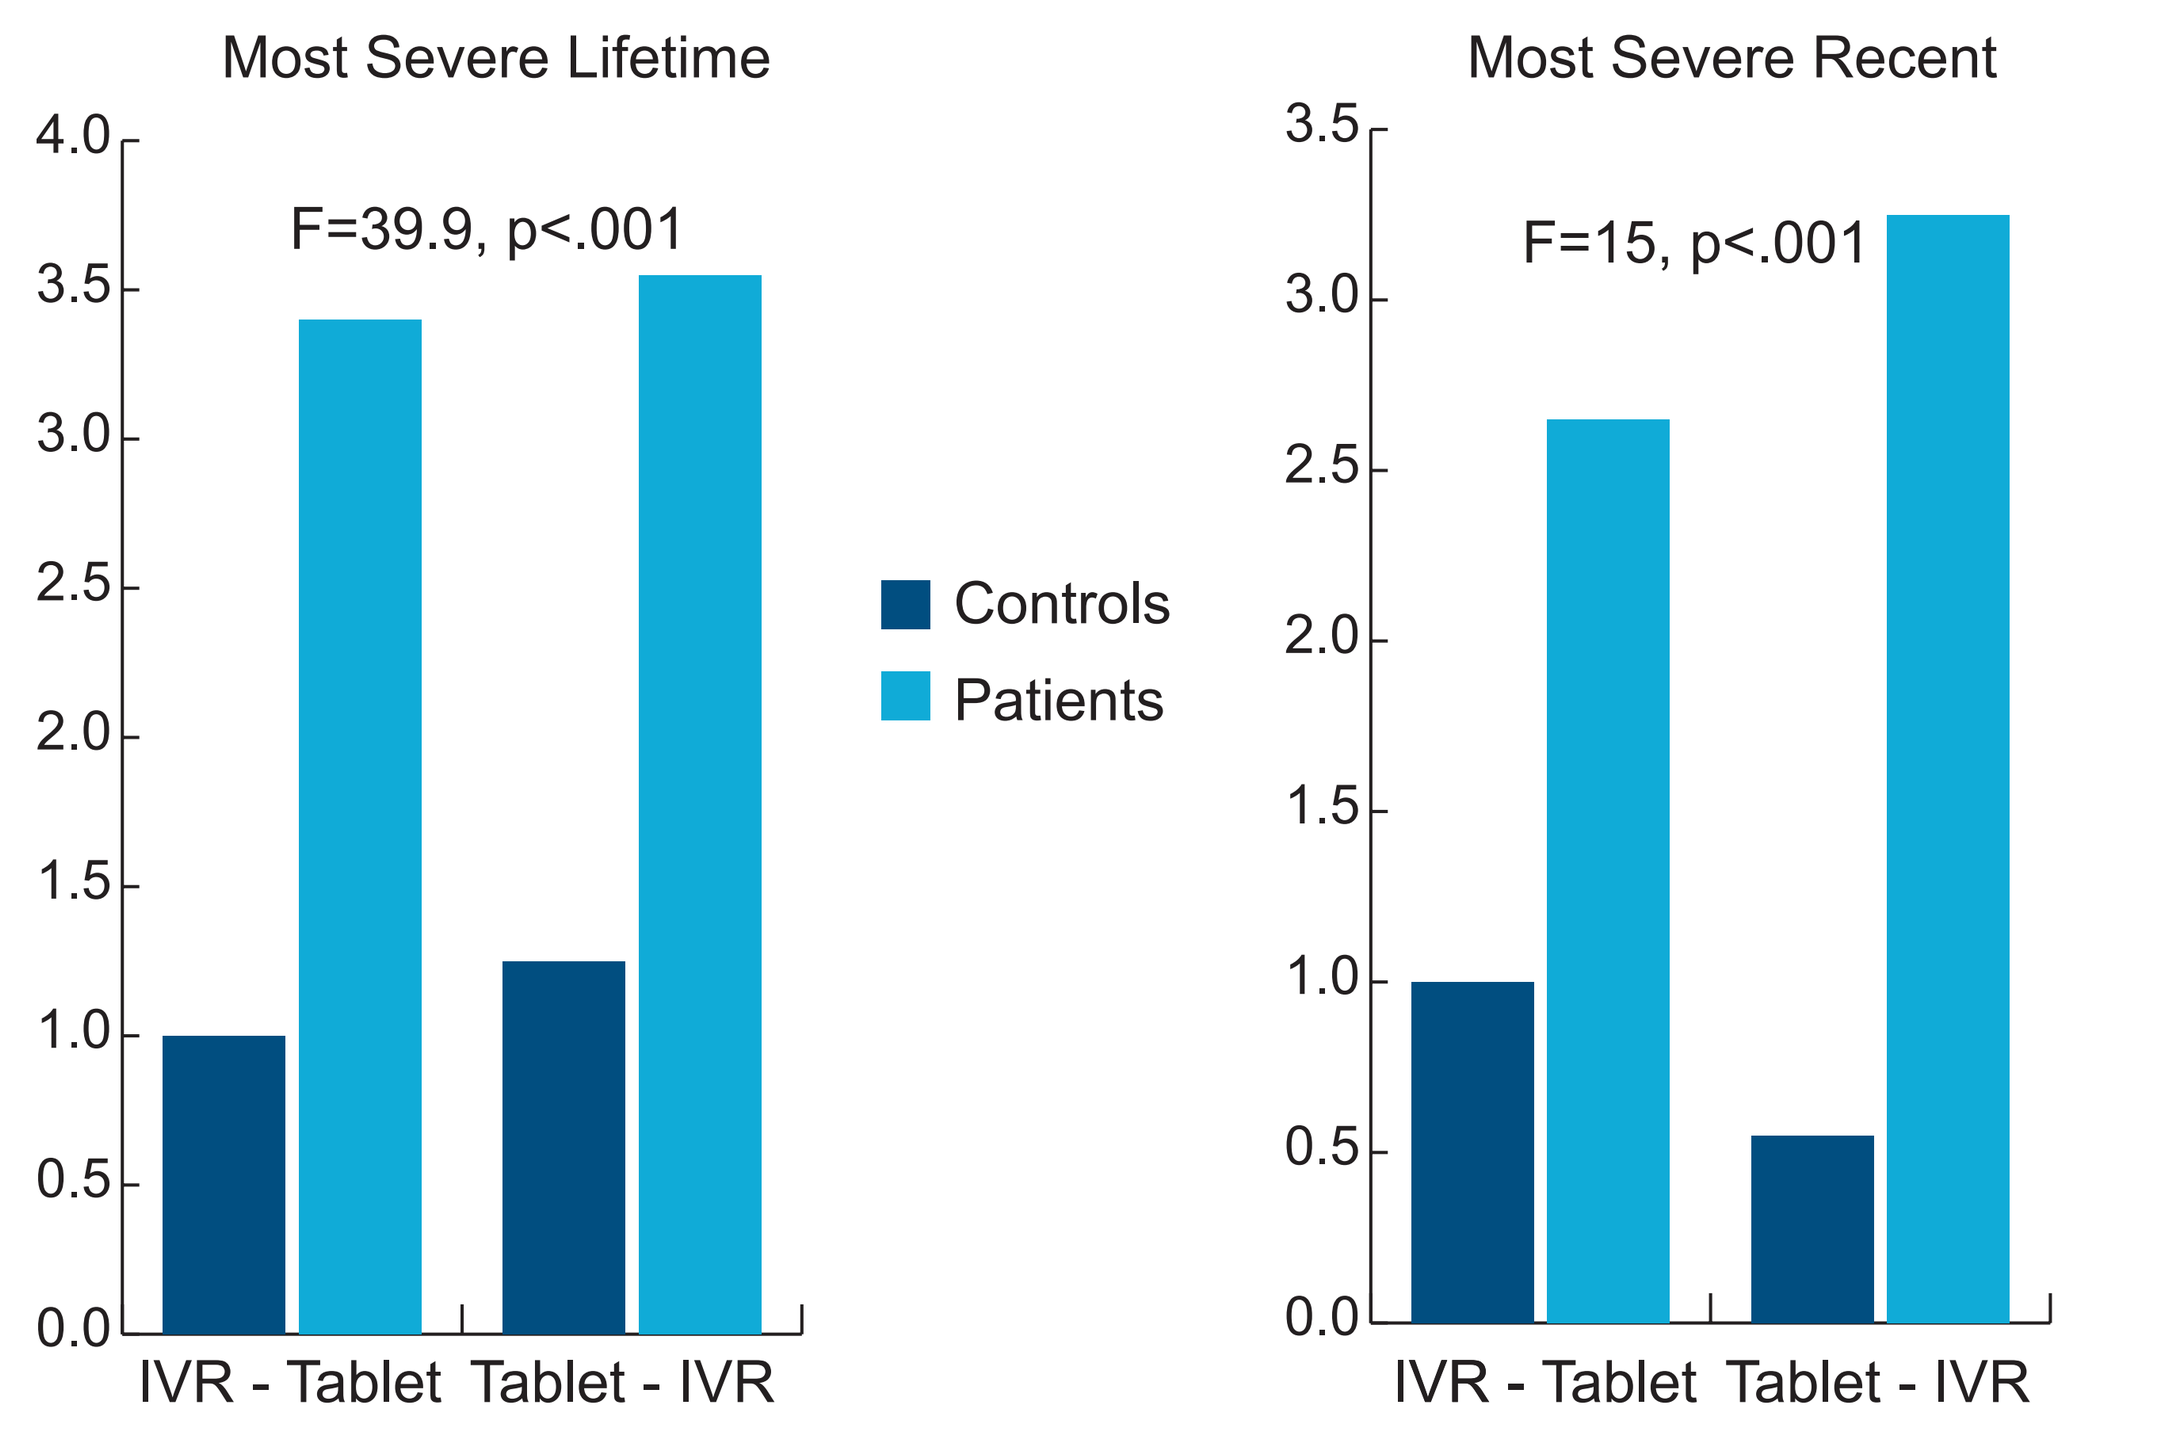
Most Severe Lifetime
F=39.9, p<.001
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
IVR - Tablet
Tablet - IVR
Controls
Patients
Most Severe Recent
F=15, p<.001
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
IVR - Tablet
Tablet - IVR

Convergence Analyses - Behavior

- Measures of equivalence all above threshold for convergence, with exception of lifetime aborted attempts
- Patients report more suicidal behavior than controls

Variable	Equivalence Kappa/Tau b	Known Groups Validity
Actual Attempts (lifetime)	.81, p<.001	X2=14.43; p. < .001
Actual Attempts (recent - last 2 years)	.73, p<.001	No controls reported recent behavior – no test possible
Number of lifetime actual attempts	.81, p<.001	t(84) = 3.36; p. = .001
Interrupted attempts (lifetime)	.78, p<.001	X2=18.22; p. < .001
Interrupted attempts (recent - last 2 years)	.62, p<.001	X2=18.22; p. < .001
Aborted attempts (lifetime)	.54, p<.001	X2=18.22; p. < .001
Aborted Attempts (recent - last 2 years)	.74, p<.001	X2=2.21; p. = .137 (only 4 cases in the control group)
Preparatory behaviors (lifetime)	.77, p<.001	X2=10.56; p. = .001
Preparatory behaviors (recent - last 2 years)	.89, p<.001	X2=1.26; p. = .262 (only 1 case in the control group)
Non suicidal self-injurious behavior	.73, p<.001	X2=0.81; p. = .367

Known-Groups Validity - Behavior

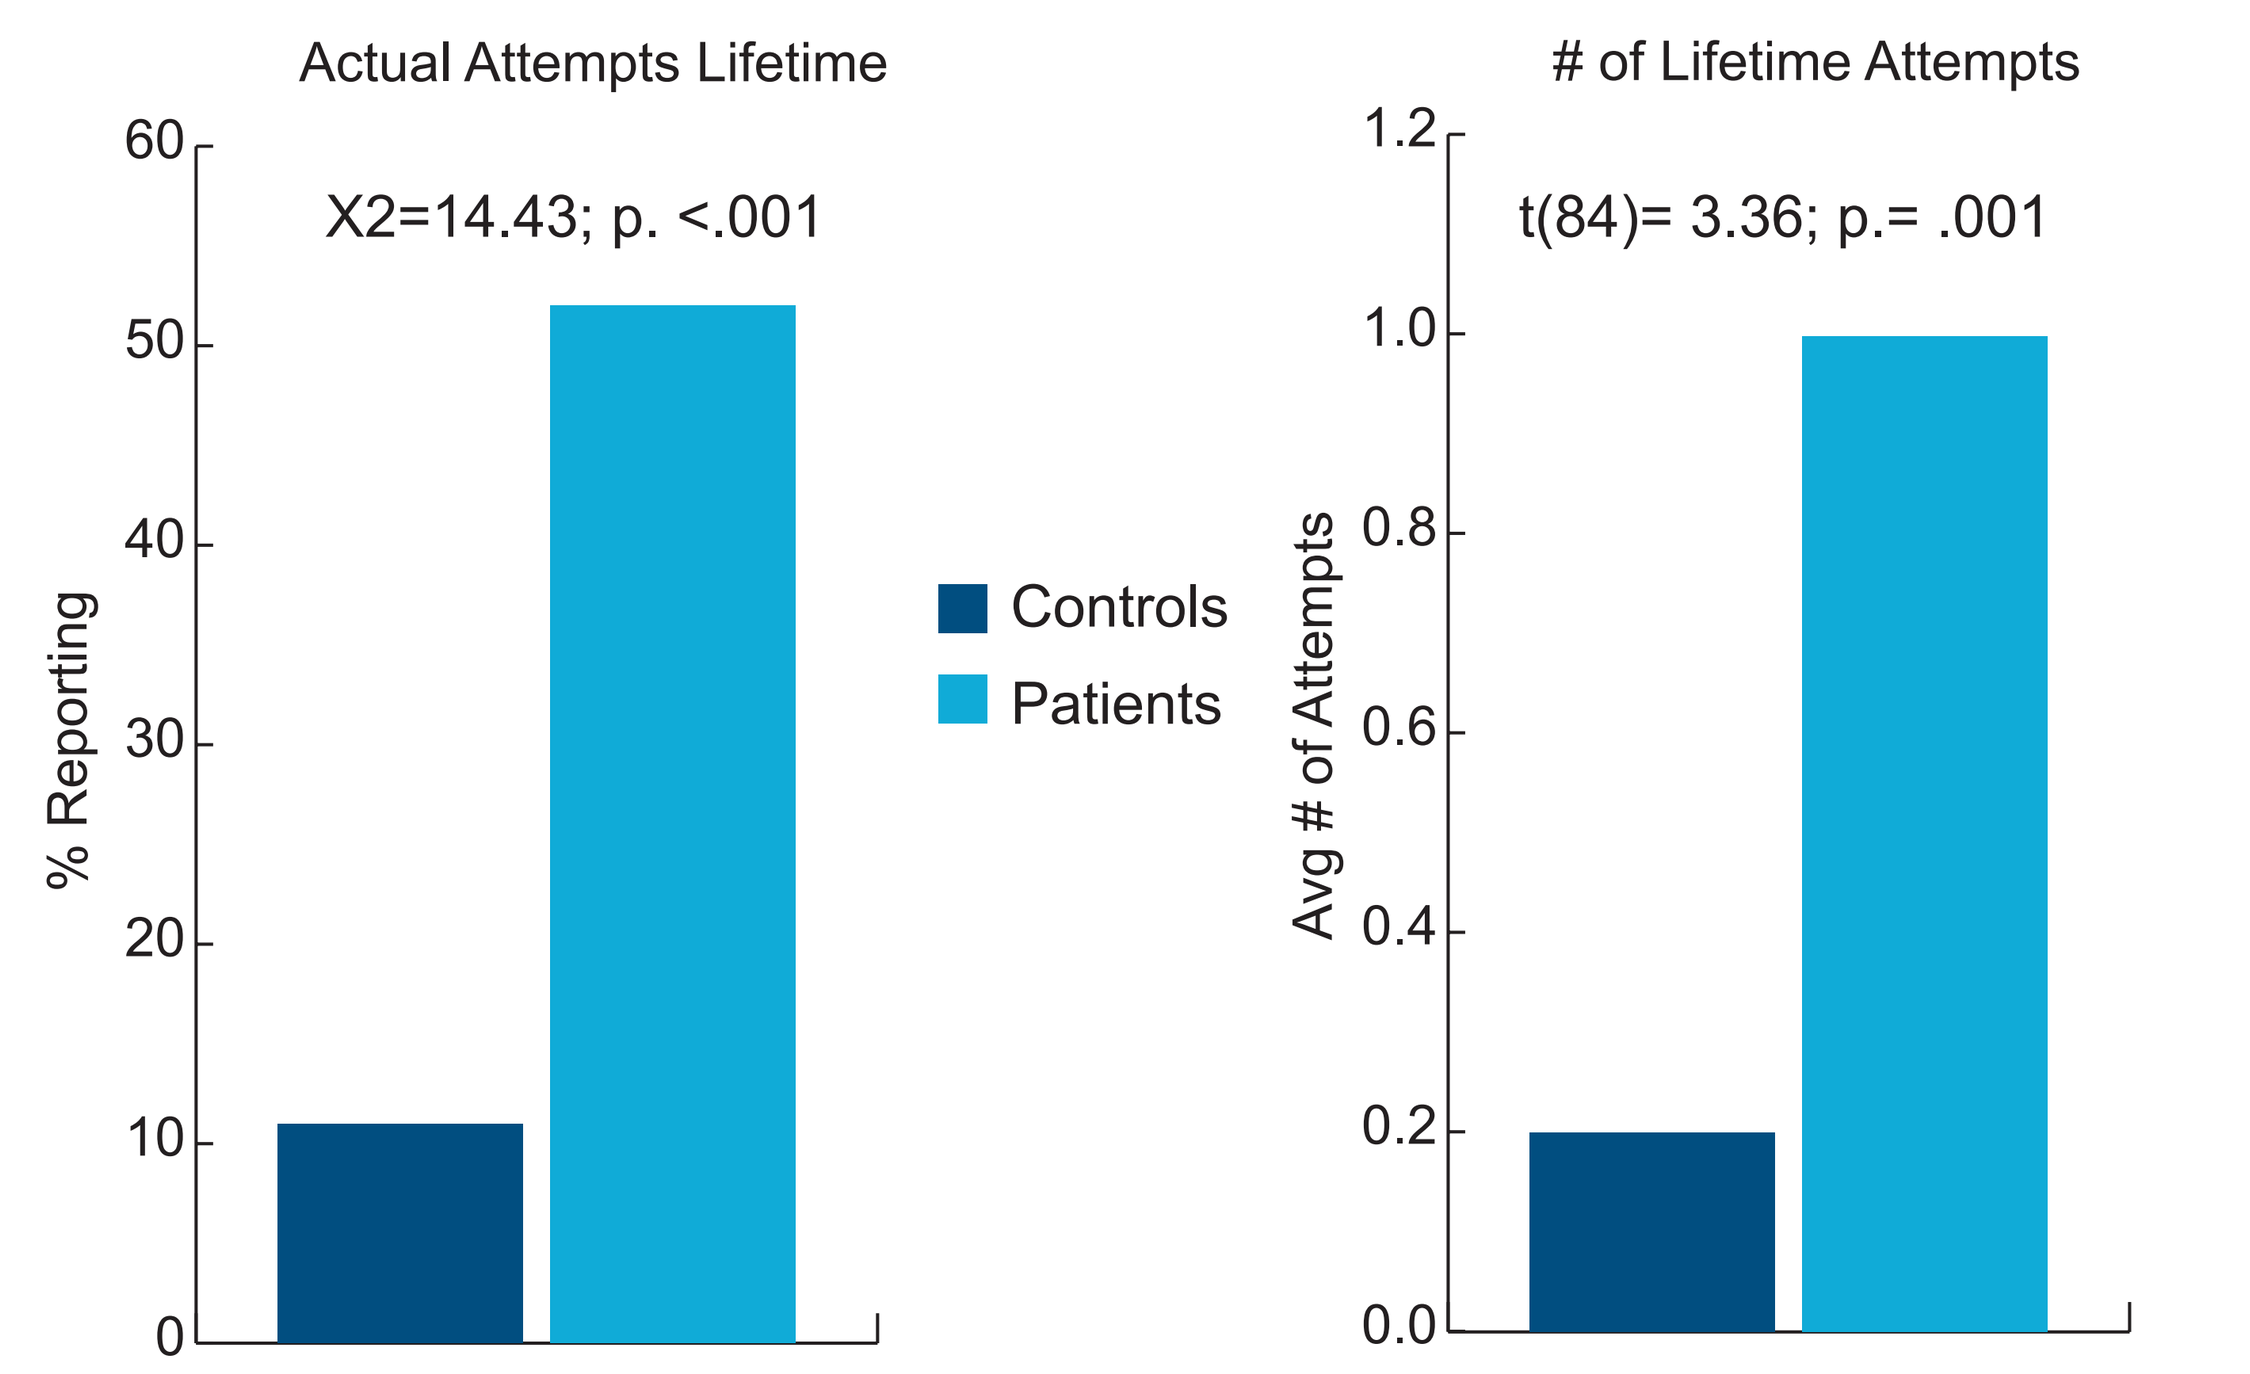
Actual Attempts Lifetime
X2=14.43; p. <.001
% Reporting
60
50
40
30
20
10
0
Controls
Patients
of Lifetime Attempts
t(84)= 3.36; p.= .001
Avg # of Attempts
1.2
1.0
0.8
0.6
0.4
0.2
0.0

Conclusions

- **IVR and Tablet versions of the eC-SSRS produced equivalent data**
- **Ideation assessments from both versions were shown to have high test-retest reliability**
- **Data from IVR and tablet versions discriminated between psychiatric inpatients and hospital worker controls**
- **Allows for the use of multiple eC-SSRS platforms in clinical research**
 - **Tablet may be particularly useful when other patient-reported outcomes are administered via tablet at the clinical site**

References

1. Food and Drug Administration (2012). Suicidal Ideation and Behavior: Prospective Assessment of Occurrence in Clinical Trials. http://www.fda.gov/drugs/guidancecomplianceregulatoryinformation/guidances/ucm315156.htm.
2. Mundt JC, Greist JH, Gelenberg AJ, Katzelnick DJ, Jefferson JW, Modell JG. (2010) Feasibility and validation of a computer-automated Columbia-Suicide severity rating scale using interactive voice response technology. J Psych Res 44:1224–1228.
3. Posner K, Brown GK, Stanley B, Brent DA, Yershova KV, Oquendo MA, Currier GW, Melvin GA, Greenhill L, Shen S, Mann JJ. (2011) The Columbia-Suicide Severity Rating Scale: internal validity and internal consistency findings from three multisite studies with adolescents and adults. Am J Psych 168:1266–1277.

Healthcare Technology Systems
Helping Technology Help People

Presented at the ISCTM 11th Annual Meeting, 19FEB2015, Washington, D.C.

ERT
Getting It Done. Right.
www.ert.com